An Evaluation of Adverse Skin Reactions in Patients with Multiple Sclerosis on Disease-Modifying Therapies

P787

K Beer,[1] M Müller,[2] A Hew-Winzeler,[3] A Bont,[4] P Maire,[5] X You,[6] P Foulds,[6] D Curtius[7]

[1]FMH Neurologie, Wil SG, Switzerland; [2]FMH Neurologie, Kantonsspital Luzern, Luzern, Switzerland; [3]FMH Neurologie, Zurich, Switzerland; [4]FMH Neurologie, Winterthur, Switzerland; [5]FMH Neurologie, Hirslanden Klinik Aarau, Aarau, Switzerland; [6]Biogen Idec, Wellesley, MA, USA; [7]Biogen-Dompe AG, Zug, Switzerland

25th Congress of the European Committee for Treatment and Research in Multiple Sclerosis

September 9–12, 2009
Düsseldorf, Germany

Background

- Interferon beta (IFNβ) and glatiramer acetate (GA) are approved therapies for the long-term treatment of multiple sclerosis (MS).
- IFNβ and GA are administered by either subcutaneous (SC) or intramuscular (IM) injection.
- Experience with MS treatments in general practice shows that patients often discontinue treatment in the first 6–12 months or do not consistently comply with it.[1]
- Adverse skin reactions are often reported by patients with MS as a reason for noncompliance or switching therapies.[2]
- Patients continue to experience adverse skin reactions after years of treatment.

Objective

- To evaluate the proportion of patients on IFNβ or GA who experience injection-site reactions (ISRs) and who switch or discontinue therapy because of adverse skin reactions.

Methods

- The Swiss MS Skin Project (SMSP) was an observational, multicenter study.
- Patients with relapsing MS or clinically isolated syndrome who had been on the same disease-modifying therapy (DMT) for some time during the past 2 years were enrolled.
- Baseline demographic information was collected and a skin examination was conducted at the screening visit.
- A follow-up examination was performed 1 year later.

Results

Patients

- A total of 499 patients were enrolled; 412 met per-protocol entry criteria were currently on 1 of 4 DMTs: IM IFNβ-1a (Avonex®, n = 82), SC IFNβ-1b (Betaferon®, n = 123), SC IFNβ-1a (Rebif®, n = 184), or GA (Copaxone®, n = 23).
- The baseline demographic and disease characteristics of patients at enrollment are summarized in Table 1.
 - Overall mean age at enrollment was 44.3 years (standard deviation [SD] 10.12).
 - The majority of patients (69.7%) were women.
 - Mean duration of MS symptoms was 9.3 years (SD 6.22).
 - Mean duration of treatment at screening was 5.9 years (SD 2.97).

Table 1. Baseline Demographic and Disease Characteristics of Patients

	IM IFNβ-1a (N = 82)	SC IFNβ-1b (N = 123)	SC IFNβ-1a (N = 184)	SC GA (N = 23)	Overall (N = 412)
Median age, years (min, max)	43.9 (23.6, 77.1)	45.3 (21.1, 66.5)	43.4 (17.1, 67.2)	44.2 (23.6, 62.5)	44.0 (17.1, 71.1)
Gender Male (%)	18 (22.0)	37 (30.6)	61 (33.3)	8 (34.8)	124 (30.3)
Gender Female (%)	64 (78.0)	84 (69.4)	122 (66.7)	15 (65.2)	285 (69.7)
Mean duration of MS disease, years (SD)	9.9 (7.37)	10.0 (6.29)	8.6 (5.74)	9.5 (4.92)	9.3 (6.22)
Median duration of MS disease, years (min, max)	8.0 (0.0, 42.1)	8.6 (2.0, 32.3)	7.0 (0.1, 26.7)	8.9 (2.3, 20.4)	7.7 (0.0, 42.1)
Mean duration of treatment, years (SD)	5.5 (2.56)	6.8 (3.06)	5.5 (2.93)	5.6 (3.24)	5.9 (2.97)
Median duration of treatment, years (min, max)	5.2 (2.0, 19.0)	6.8 (2.0, 13.0)	4.9 (2.0, 27.2)	5.0 (2.0, 16.9)	5.4 (2.0, 27.2)

Skin Examinations

Baseline

- Significantly fewer patients on IM IFNβ-1a (13.4%) reported ISRs compared with patients on IFNβ-1b (57.7%, $P < .0001$), patients on SC IFNβ-1a (67.9%, $P < .0001$), and patients on GA (30.4%; $P = .056$) (Figure 1).

Figure 1. Skin Status at Baseline

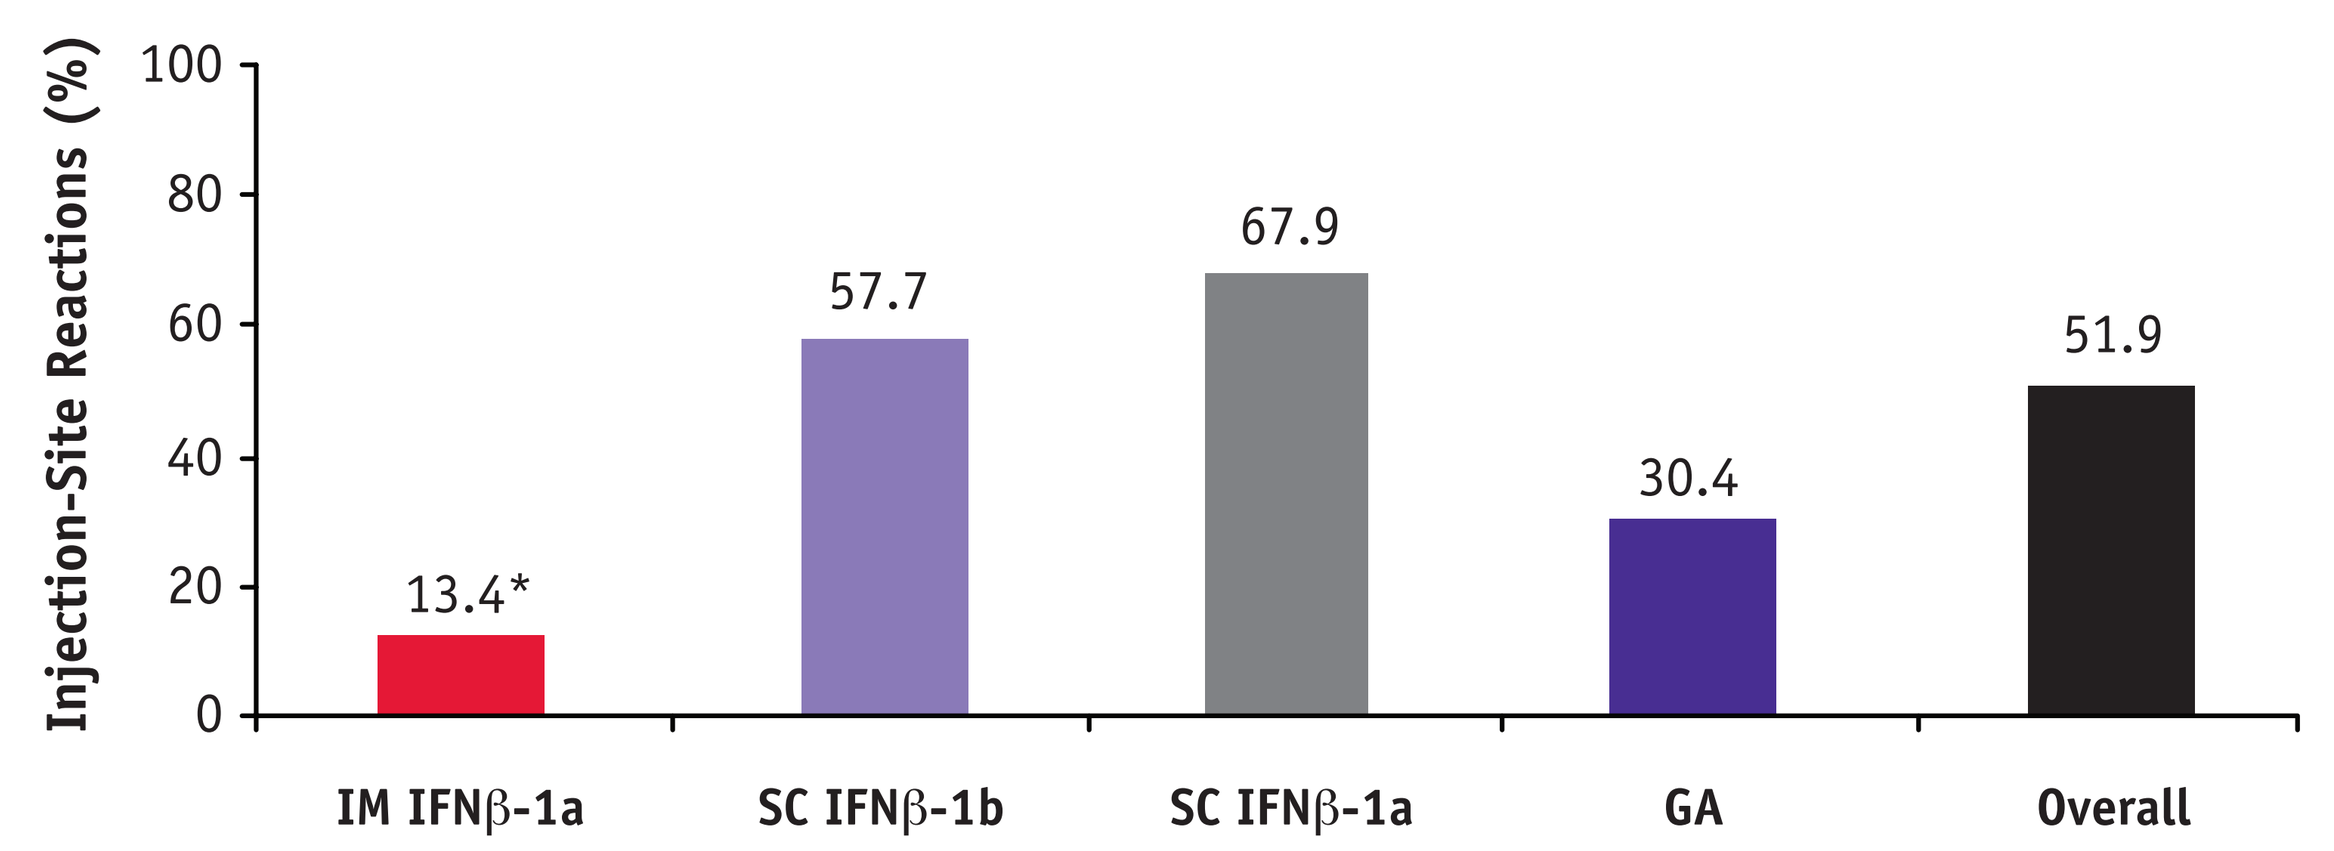
Injection-Site Reactions (%)
100
80
60
40
20
0
13.4*
57.7
67.9
30.4
51.9
IM IFNβ-1a
SC IFNβ-1b
SC IFNβ-1a
GA
Overall

***$P < .0001$ vs SC IFNβ-1b and SC IFNβ-1a; $P = .056$ vs SC GA.**

- Patients on IM IFNβ-1a were significantly less likely to have necrosis or lipoatrophy (0; 1.2%) compared with patients on SC IFNβ-1b (5.7%; 8.9%) or SC IFNβ-1a (6.0%; 10.3%) (Table 2).

Table 2. Evaluation of Skin Status at Baseline

	IM IFNβ-1a (N = 82)	SC IFNβ-1b (N = 123)	SC IFNβ-1a (N = 184)	SC GA (N = 23)	Overall (N = 412)
Necrosis, N (%)	0 (0)[a]	7 (5.7)	11 (6.0)	0 (0)	18 (4.4)
Lipoatrophy, N (%)	1 (1.2)[b]	11 (8.9)	19 (10.3)	3 (13.0)	34 (8.3)
Injection omitted during last 4 weeks, N (%)	0 (0)[c]	7 (5.7)	13 (7.1)	1 (4.3)	21 (5.1)

[a]$P = .0279$ vs SC IFNβ-1b; $P = .0201$ vs SC IFNβ-1a.
[b]$P = .0210$ vs SC IFNβ-1b; $P = .0093$ vs SC IFNβ-1a; $P = .0322$ vs SC GA.
[c]$P = .0435$ vs SC IFNβ-1b; $P = .0114$ vs SC IFNβ-1a.

- Patients on GA were significantly more likely to have lipoatrophy compared with patients on IM IFNβ-1a (13% vs 1.2%, $P = .032$) (Table 2).
- No patient on IM IFNβ-1a had missed a dose in the previous 4 weeks because of an ISR, compared with 5.7% of patients on SC IFNβ-1b ($P = .044$), 7.1% of patients on SC IFNβ-1a ($P = .011$), and 4.3% of patients on GA (P = NS).

1-Year Follow-up

- A significantly higher proportion of patients originally on IM IFNβ-1a were still on the same therapy compared with patients who started on other DMTs (86.6%, overall $P = .0364$) (Figure 2).

Figure 2. Patients Remaining on Original Treatment at 1-Year Follow-up

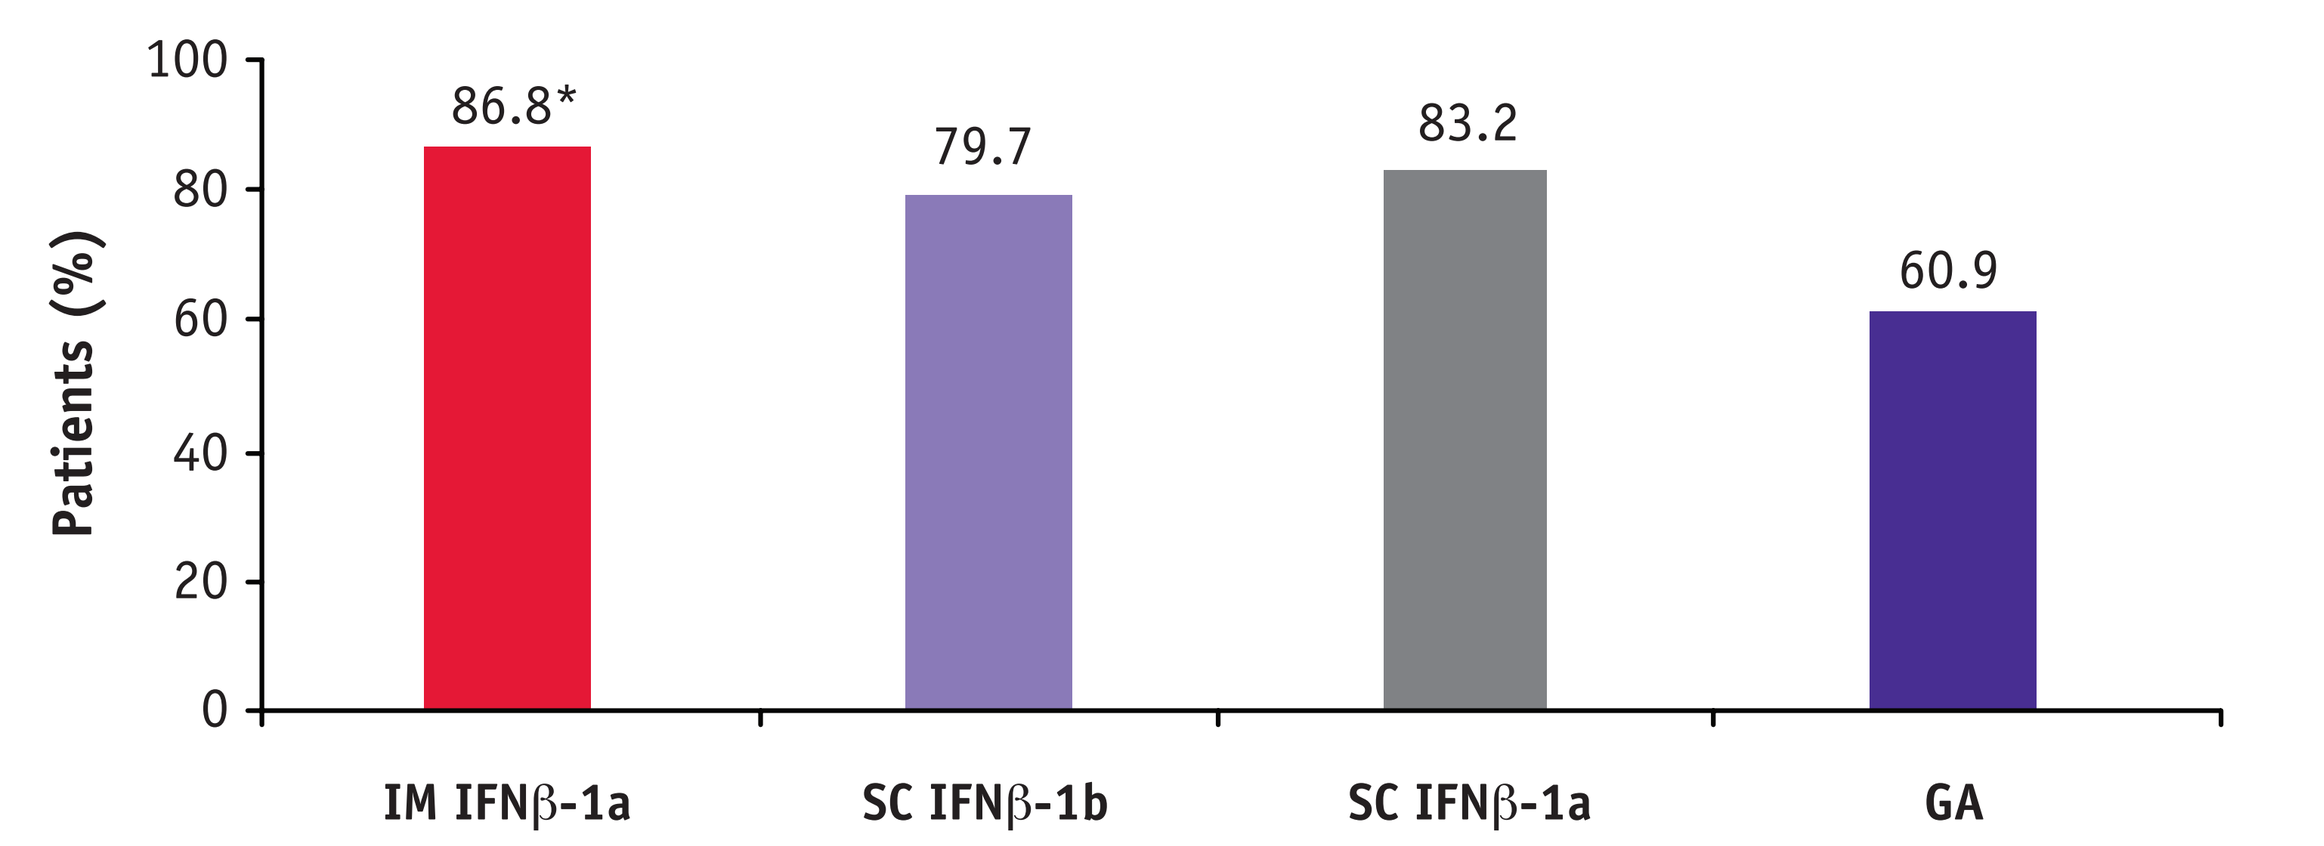
Patients (%)
100
80
60
40
20
0
86.8*
79.7
83.2
60.9
IM IFNβ-1a
SC IFNβ-1b
SC IFNβ-1a
GA

***Overall P (chi-square test) = .0364.**

- Patients on IM IFNβ-1a were less likely to have necrosis or lipoatrophy (0;0) than patients on SC IFNβ-1b (4.8%; 8.7% [$P = .0099$]) or SC IFNβ-1a (3.2%; 15% [$P = .0099$]) at the 1-year follow-up (Table 3).

Table 3. Evaluation of Skin Status at 1-Year Follow-up

	IM IFNβ-1a (N = 73)	SC IFNβ-1b (N = 104)	SC IFNβ-1a (N = 154)	SC GA (N = 14)	Overall (N = 347)
Necrosis, N (%)	0 (0)[a]	5 (4.8)	5 (3.2)	0 (0)	10 (2.9)
Lipoatrophy, N (%)	0 (0)[b]	9 (8.7)	23 (15.0)	1 (7.1)	33 (9.6)

[a]$P = .0574$ vs SC IFNβ-1b.
[b]$P = .0099$ vs SC IFNβ-1b; $P = .0005$ vs SC IFNβ-1a.

Conclusions

- This study was conducted on a very large dataset; 499 patients were enrolled with 412 assessed per protocol.
- Significantly fewer IM IFNβ-1a patients experienced ISRs compared with other DMTs at baseline.
 - Significantly fewer IM IFNβ-1a patients experienced necrosis compared with patients on other IFN formulations.
 - Significantly fewer IM IFNβ-1a patients experienced lipoatrophy compared with patients on other DMTs.
 - No IM IFNβ-1a patients skipped a dose in previous 4 weeks because of ISRs, unlike the other DMTs; significantly more patients skipped a dose of the other IFN formulations.
- Similar patterns for necrosis and lipoatrophy rates were seen at the 1-year follow-up.
 - Significantly fewer IM IFNβ-1a patients experienced necrosis compared with SC IFNβ-1b patients.
 - Significantly fewer IM IFNβ-1a patients experienced lipoatrophy compared with patients on other IFN formulations.
- More patients remained on IM IFNβ-1a compared with other DMTs over the 1-year study.
- Patients on IM IFNβ-1a had fewer skin reactions and better compliance and were less likely to switch to other DMTs compared with patients on other therapies.
- Minimizing the impact of adverse effects is crucial in helping patients adhere to their treatment regimens and in improving their chances of better health over the longer term.[2]

References

1. Tremlett HL, Oger J. *Neurology.* 2003;61:551–554.
2. Girouard N, Théorêt G. *Can J Neurosci Nurs.* 2008;30:18–25.

Study supported by Biogen Idec.

An Evaluation of Adverse Skin Reactio
on Disease-Mod

P787

K Beer,[1] M Müller,[2] A Hew-Winzeler,[3] A Bo

[1]FMH Neurologie, Wil SG, Switzerland; [2]FMH Neurologie, Kantonsspital Luzern, Luzern, Swi
[5]FMH Neurologie, Hirslanden Klinik Aarau, Aarau, Switzerland; [6]Bio

Background

- Interferon beta (IFNβ) and glatiramer acetate (GA) are approved therapies for the long-term treatment of multiple sclerosis (MS).
- IFNβ and GA are administered by either subcutaneous (SC) or intramuscular (IM) injection.
- Experience with MS treatments in general practice shows that patients often discontinue treatment in the first 6–12 months or do not consistently comply with it.[1]
- Adverse skin reactions are often reported by patients with MS as a reason for noncompliance or switching therapies.[2]
- Patients continue to experience adverse skin reactions after years of treatment.

Objective

- To evaluate the proportion of patients on IFNβ or GA who experience injection-site reactions (ISRs) and who switch or discontinue therapy because of adverse skin reactions.

Methods

- The Swiss MS Skin Project (SMSP) was an observational, multicenter study.
- Patients with relapsing MS or clinically isolated syndrome who had been on the same disease-modifying therapy (DMT) for some time during the past 2 years were enrolled.
- Baseline demographic information was collected and a skin examination was conducted at the screening visit.
- A follow-up examination was performed 1 year later.

Results

Patients

- A total of 499 patients were enrolled; 412 met per-protocol entry criteria were currently on 1 of 4 DMTs: IM IFNβ-1a (Avonex®, n = 82), SC IFNβ-1b (Betaferon®, n = 123), SC IFNβ-1a (Rebif®, n = 184), or GA (Copaxone®, n = 23).
- The baseline demographic and disease characteristics of patients at enrollment are summarized in Table 1.
 - Overall mean age at enrollment was 44.3 years (standard deviation [SD] 10.12).
 - The majority of patients (69.7%) were women.
 - Mean duration of MS symptoms was 9.3 years (SD 6.22).
 - Mean duration of treatment at screening was 5.9 years (SD 2.97).

Table 1. Baseline Demographic and Disease Characteristics of Patients

	IM IFNβ-1a (N = 82)	SC IFNβ-1b (N = 123)	SC IFNβ-1a (N = 184)	SC GA (N = 23)	Overall (N = 412)
Median age, years (min, max)	43.9 (23.6, 77.1)	45.3 (21.1, 66.5)	43.4 (17.1, 67.2)	44.2 (23.6, 62.5)	44.0 (17.1, 71.1)
Gender Male (%)	18 (22.0)	37 (30.6)	61 (33.3)	8 (34.8)	124 (30.3)
Gender Female (%)	64 (78.0)	84 (69.4)	122 (66.7)	15 (65.2)	285 (69.7)
Mean duration of MS disease, years (SD)	9.9 (7.37)	10.0 (6.29)	8.6 (5.74)	9.5 (4.92)	9.3 (6.22)
Median duration of MS disease, years (min, max)	8.0 (0.0, 42.1)	8.6 (2.0, 32.3)	7.0 (0.1, 26.7)	8.9 (2.3, 20.4)	7.7 (0.0, 42.1)
Mean duration of treatment, years (SD)	5.5 (2.56)	6.8 (3.06)	5.5 (2.93)	5.6 (3.24)	5.9 (2.97)
Median duration of treatment, years (min, max)	5.2 (2.0, 19.0)	6.8 (2.0, 13.0)	4.9 (2.0, 27.2)	5.0 (2.0, 16.9)	5.4 (2.0, 27.2)

Skin Examinations

Baseline

- Significantly fewer patients on IM IFNβ-1a (13.4%) reported ISRs compared with patients on IFNβ-1b (57.7%, $P < .0001$), patients on SC IFNβ-1a (67.9%, $P < .0001$), and patients on GA (30.4%; $P = .056$) (Figure 1).

Figure 1. Skin Status at Baseline

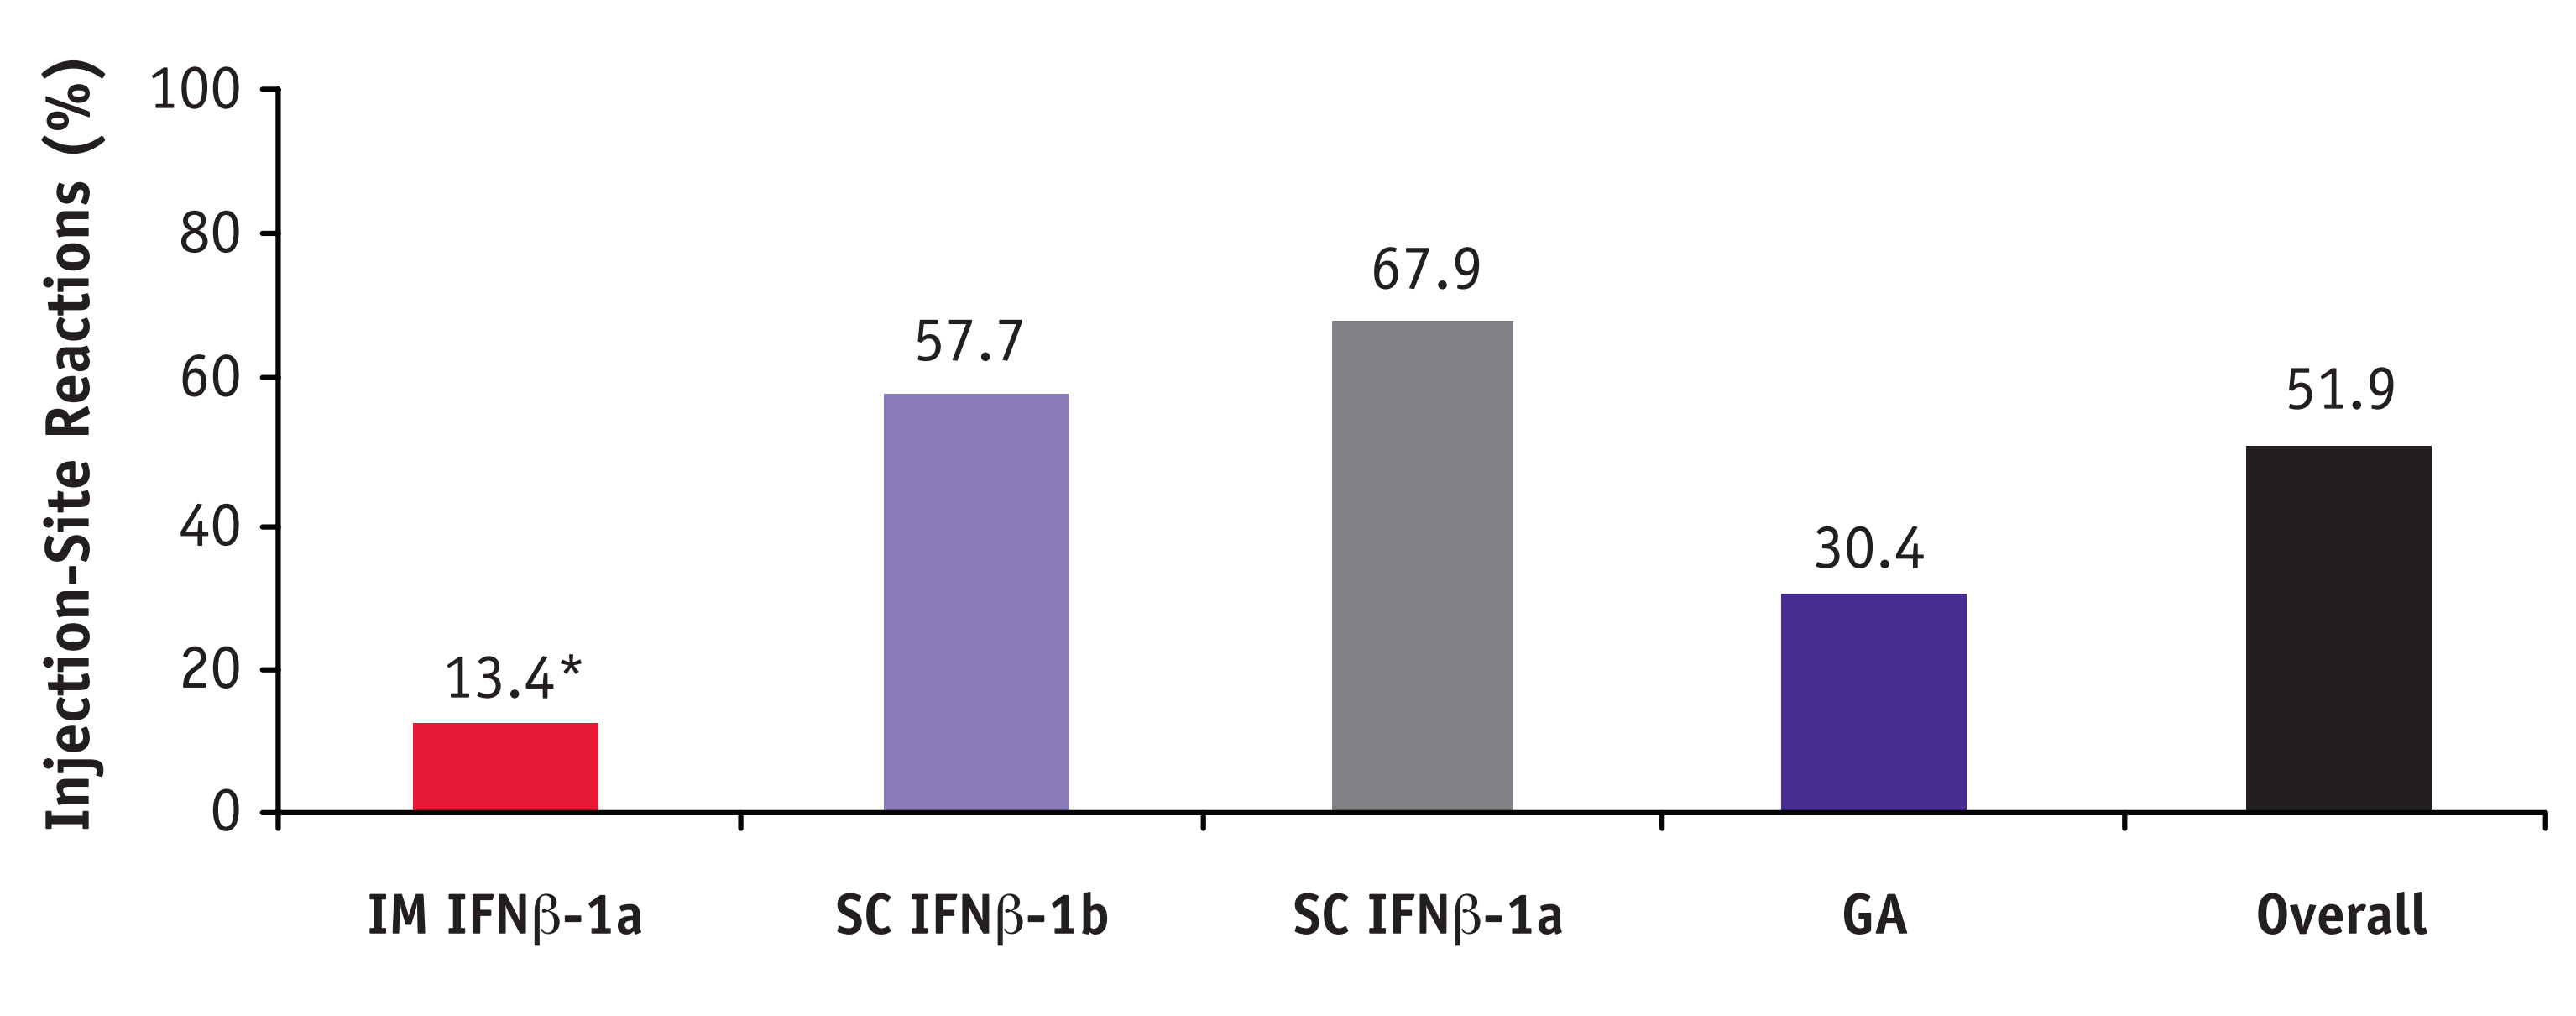
Injection-Site Reactions (%)
100
80
60
40
20
0
13.4*
57.7
67.9
30.4
51.9
IM IFNβ-1a
SC IFNβ-1b
SC IFNβ-1a
GA
Overall

******P* < .0001 vs SC IFNβ-1b and SC IFNβ-1a; *P* = .056 vs SC GA.**

- Patients on IM IFNβ-1a were significantly less likely to have necrosis or lipoatrophy (0; 1.2%) compared with patients on SC IFNβ-1b (5.7%; 8.9%) or SC IFNβ-1a (6.0%; 10.3%) (Table 2).

ons in Patients with Multiple Sclerosis
ifying Therapies

nt,[4] P Maire,[5] X You,[6] P Foulds,[6] D Curtius[7]

itzerland; [3]FMH Neurologie, Zurich, Switzerland; [4]FMH Neurologie, Winterthur, Switzerland;
gen Idec, Wellesley, MA, USA; [7]Biogen-Dompe AG, Zug, Switzerland

25th Congress of the European Committee for Treatment and Research in Multiple Sclerosis

September 9–12, 2009
Düsseldorf, Germany

Table 2. Evaluation of Skin Status at Baseline

	IM IFNβ-1a (N = 82)	SC IFNβ-1b (N = 123)	SC IFNβ-1a (N = 184)	SC GA (N = 23)	Overall (N = 412)
Necrosis, N (%)	0 (0)[a]	7 (5.7)	11 (6.0)	0 (0)	18 (4.4)
Lipoatrophy, N (%)	1 (1.2)[b]	11 (8.9)	19 (10.3)	3 (13.0)	34 (8.3)
Injection omitted during last 4 weeks, N (%)	0 (0)[c]	7 (5.7)	13 (7.1)	1 (4.3)	21 (5.1)

[a]*P* = .0279 vs SC IFNβ-1b; *P* = .0201 vs SC IFNβ-1a.
[b]*P* = .0210 vs SC IFNβ-1b; *P* = .0093 vs SC IFNβ-1a; *P* = .0322 vs SC GA.
[c]*P* = .0435 vs SC IFNβ-1b; *P* = .0114 vs SC IFNβ-1a.

- Patients on GA were significantly more likely to have lipoatrophy compared with patients on IM IFNβ-1a (13% vs 1.2%, *P* = .032) (Table 2).
- No patient on IM IFNβ-1a had missed a dose in the previous 4 weeks because of an ISR, compared with 5.7% of patients on SC IFNβ-1b (*P* = .044), 7.1% of patients on SC IFNβ-1a (*P* = .011), and 4.3% of patients on GA (*P* = NS).

1-Year Follow-up

- A significantly higher proportion of patients originally on IM IFNβ-1a were still on the same therapy compared with patients who started on other DMTs (86.6%, overall *P* = .0364) (Figure 2).

Figure 2. Patients Remaining on Original Treatment at 1-Year Follow-up

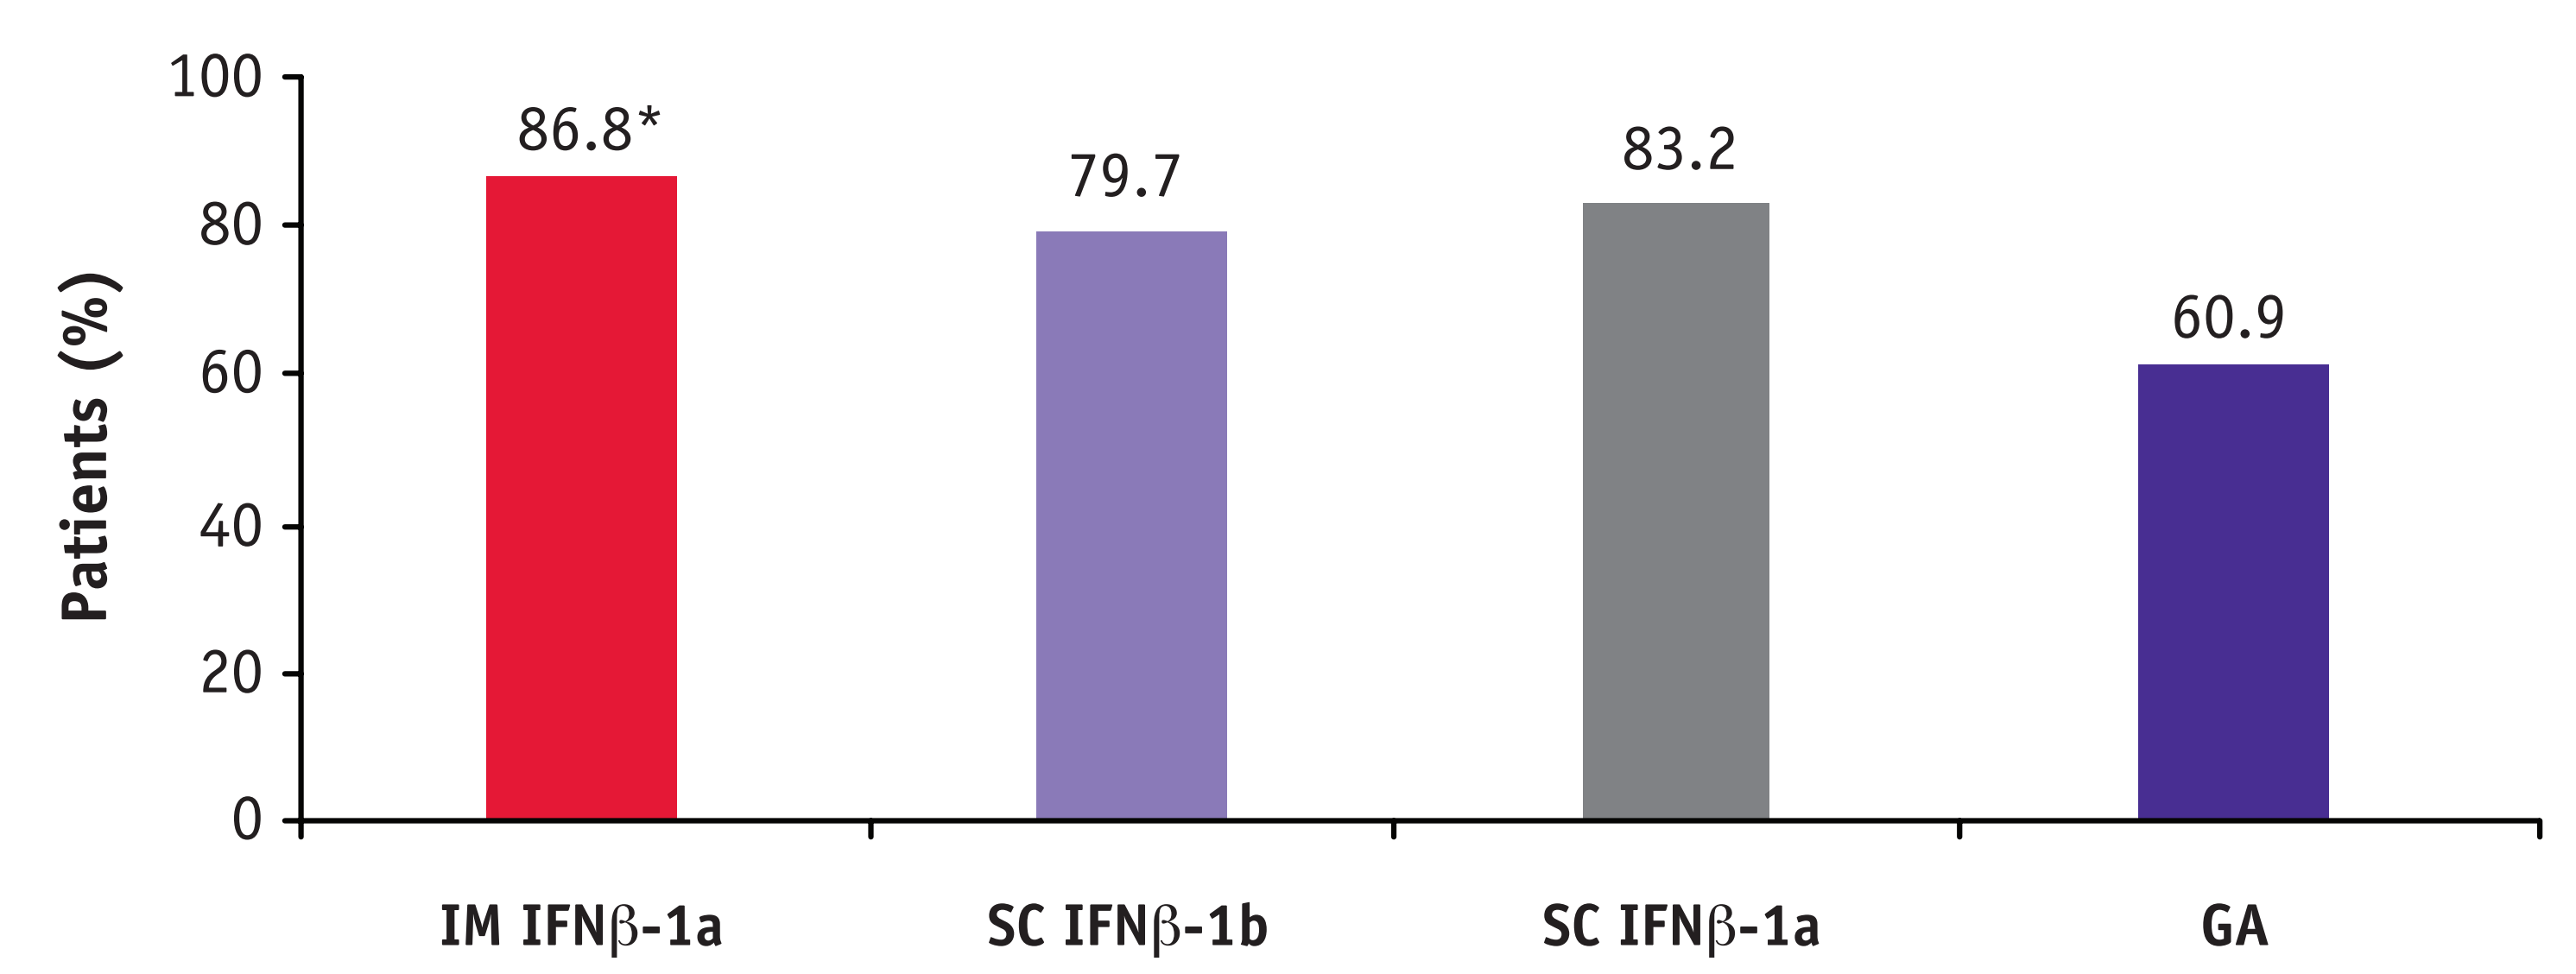
Patients (%)
100
80
60
40
20
0
86.8*
79.7
83.2
60.9
IM IFNβ-1a
SC IFNβ-1b
SC IFNβ-1a
GA

***Overall *P* (chi-square test) = .0364.**

- Patients on IM IFNβ-1a were less likely to have necrosis or lipoatrophy (0;0) than patients on SC IFNβ-1b (4.8%; 8.7% [*P* = .0099]) or SC IFNβ-1a (3.2%; 15% [*P* = .0099]) at the 1-year follow-up (Table 3).

Table 3. Evaluation of Skin Status at 1-Year Follow-up

	IM IFNβ-1a (N = 73)	SC IFNβ-1b (N = 104)	SC IFNβ-1a (N = 154)	SC GA (N = 14)	Overall (N = 347)
Necrosis, N (%)	0 (0)[a]	5 (4.8)	5 (3.2)	0 (0)	10 (2.9)
Lipoatrophy, N (%)	0 (0)[b]	9 (8.7)	23 (15.0)	1 (7.1)	33 (9.6)

[a]*P* = .0574 vs SC IFNβ-1b.
[b]*P* = .0099 vs SC IFNβ-1b; *P* = .0005 vs SC IFNβ-1a.

Conclusions

- This study was conducted on a very large dataset; 499 patients were enrolled with 412 assessed per protocol.
- Significantly fewer IM IFNβ-1a patients experienced ISRs compared with other DMTs at baseline.
 - Significantly fewer IM IFNβ-1a patients experienced necrosis compared with patients on other IFN formulations.
 - Significantly fewer IM IFNβ-1a patients experienced lipoatrophy compared with patients on other DMTs.
 - No IM IFNβ-1a patients skipped a dose in previous 4 weeks because of ISRs, unlike the other DMTs; significantly more patients skipped a dose of the other IFN formulations.
- Similar patterns for necrosis and lipoatrophy rates were seen at the 1-year follow-up.
 - Significantly fewer IM IFNβ-1a patients experienced necrosis compared with SC IFNβ-1b patients.
 - Significantly fewer IM IFNβ-1a patients experienced lipoatrophy compared with patients on other IFN formulations.
- More patients remained on IM IFNβ-1a compared with other DMTs over the 1-year study.
- Patients on IM IFNβ-1a had fewer skin reactions and better compliance and were less likely to switch to other DMTs compared with patients on other therapies.
- Minimizing the impact of adverse effects is crucial in helping patients adhere to their treatment regimens and in improving their chances of better health over the longer term.[2]

References

1. Tremlett HL, Oger J. *Neurology*. 2003;61:551–554.
2. Girouard N, Théorêt G. *Can J Neurosci Nurs*. 2008;30:18–25.

Study supported by Biogen Idec.